polyNaut: a non-viral nanovesicle platform enabling cell-specific targeting for in vivo CAR-T therapies

Amrita Sikder[1], Vibhav Valsangkar[1], Leticia Tordesillas[1], Antonina Kouli[1], Thea Konstantinou[1], Matthew Hockley[1], Max Mascelloni[1], Antonia Cernischi[1], Liam Pratt[1], Patrick Vilela[1], Marius Kausas[1], Pravallika Manjappa[1], Elisabetta Bottaro[1], Virginia Gouveia[1], Sarah Wythe[1], Denis Cecchin[1], Ray Jupp[1]

1 ViaNautis Bio, Cambridge, United Kingdom

ViaNautis
TARGETED NANOMEDICINES

Introduction

Efficient targeted delivery remains a bottleneck in the development of safe, effective, redosable and scalable genetic medicines. Lipid nanoparticle (LNP) approaches have been extensively investigated, however, there are lingering concerns, namely, safety, issues with specific targeting, manufacturing complexity, immunogenicity and ultracold storage requirements.

To overcome these challenges, we developed polyNaut, a modular, disease-agnostic polymer nanovesicle platform, designed to enable a plug-and-play approach for precise targeting to specific cells and tissues, including immune cells, lung epithelial cells and the central nervous system across the blood-brain barrier.

As a proof-of-concept, we generated CD8-tPNVs and demonstrated precise CD8 positive immune cell targeting of CD8-tPNVs in vitro and in vivo. Immune cell-targeting tPNVs support stable, redosable, systemically administered in vivo CAR-T therapies, with broad applications across I&I and oncology, and the potential to provide scalable, accessible treatment options for large patient populations.

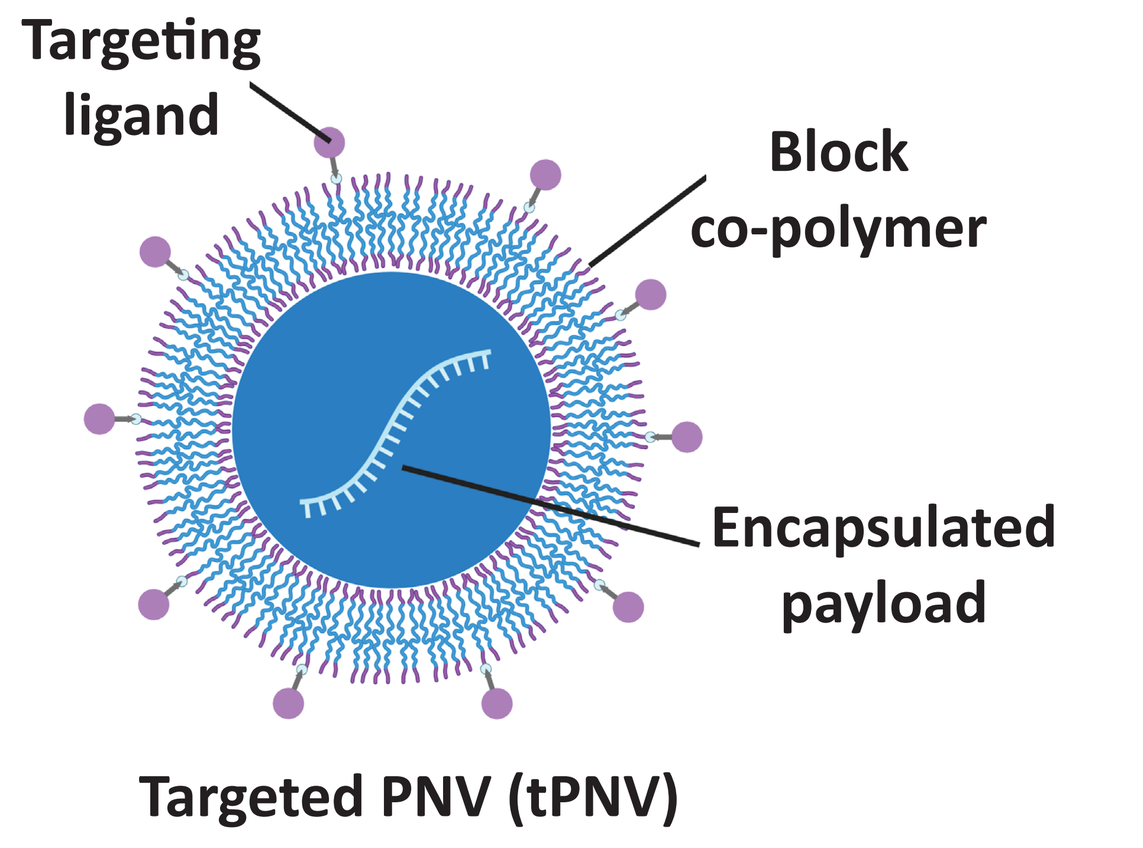
Targeting ligand
Block co-polymer
Encapsulated payload
Targeted PNV (tPNV)

Figure 1. Simple and scalable tPNV manufacturing, with targeting precision enabled by nanobody conjugation

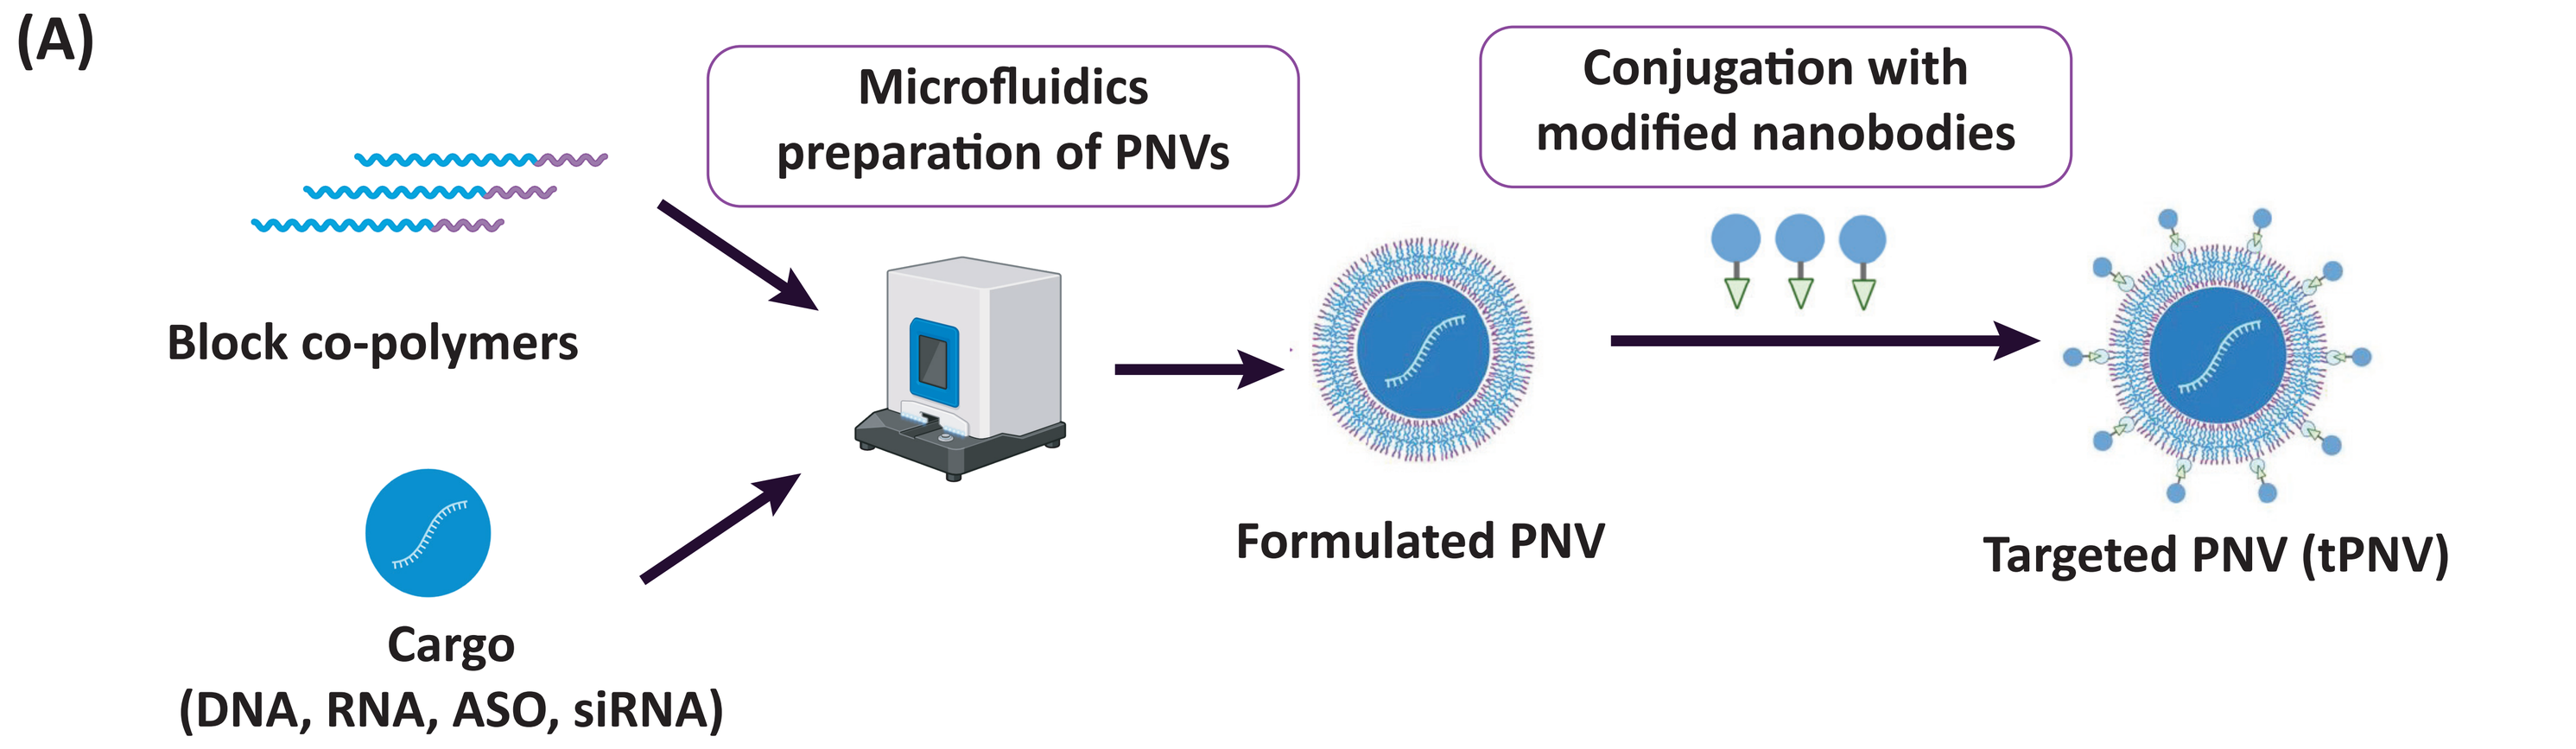
(A)
Microfluidics preparation of PNVs
Conjugation with modified nanobodies
Block co-polymers
Cargo
(DNA, RNA, ASO, siRNA)
Formulated PNV
Targeted PNV (tPNV)

(A) Schematic of the manufacturing process of generating a targeted polyNaut nanovesicle (tPNV). A tPNV is made up of three key components: a single type of block co-polymer, a cargo and a nanobody for targeting a defined cell surface receptor. The production of tPNV is automated, GMP-compatible, with each core unit operations – reaction, purification, filtration and formulation – translatable into standard CDMO infrastructure.

Figure 2. Nanobody-conjugated tPNVs exhibit high stability

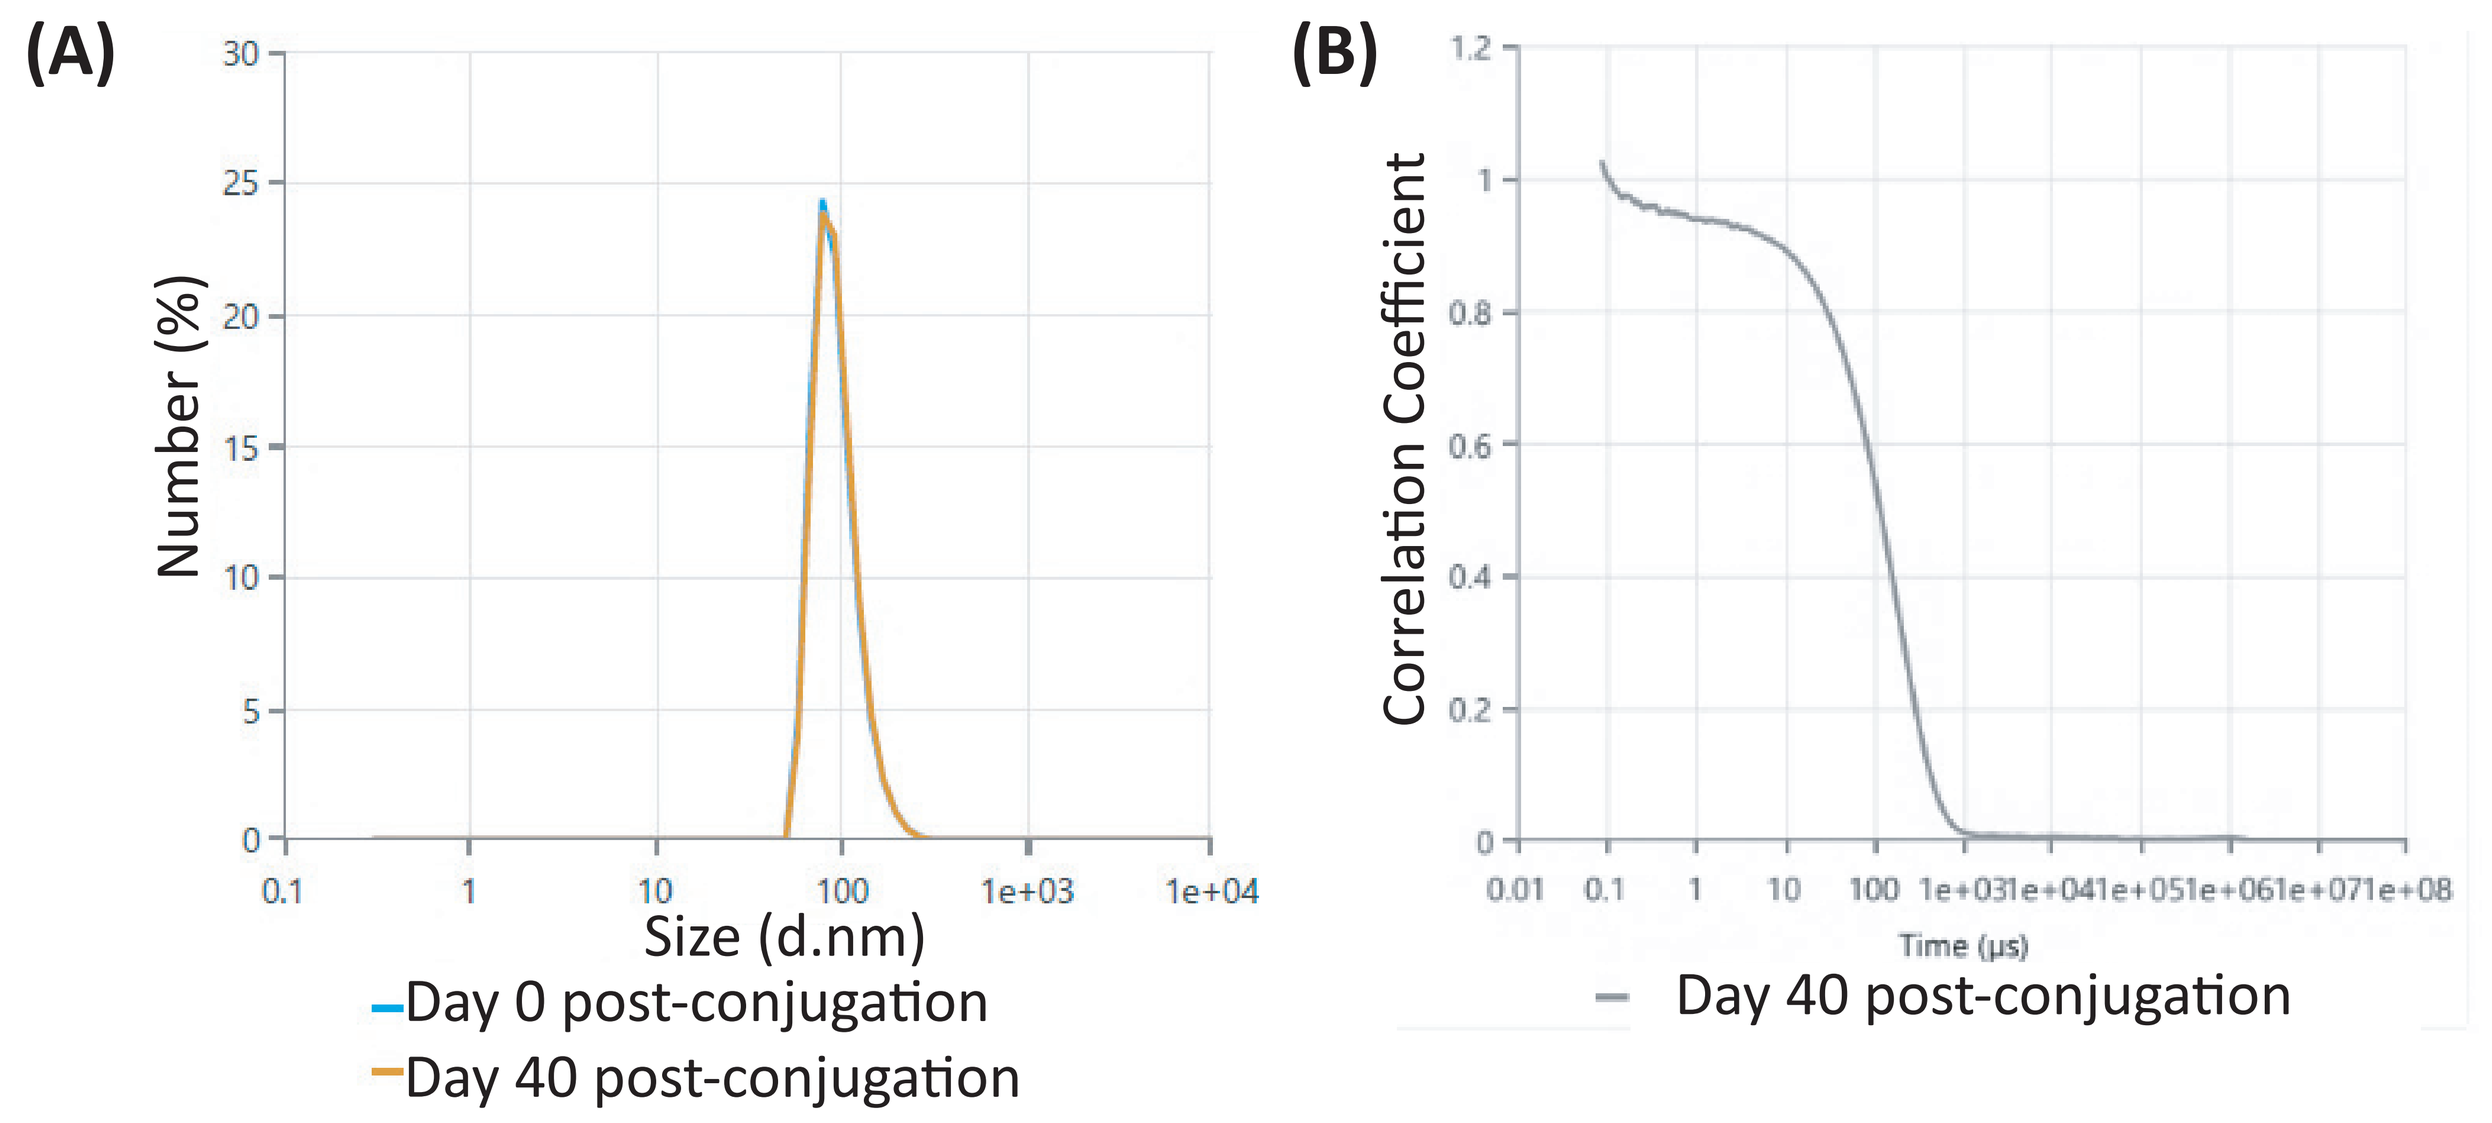
(A)
Number (%)
Size (d.nm)
Day 0 post-conjugation
Day 40 post-conjugation
(B)
Correlation Coefficient
Time (µs)
Day 40 post-conjugation

(A, B) 40 days post-conjugation, tPNVs remain stable with no change in size distribution (A) and no agglomeration (B), as measured by Dynamic Light Scattering (DLS).

Figure 3. In vitro validation of CD8-specific targeting by tPNVs

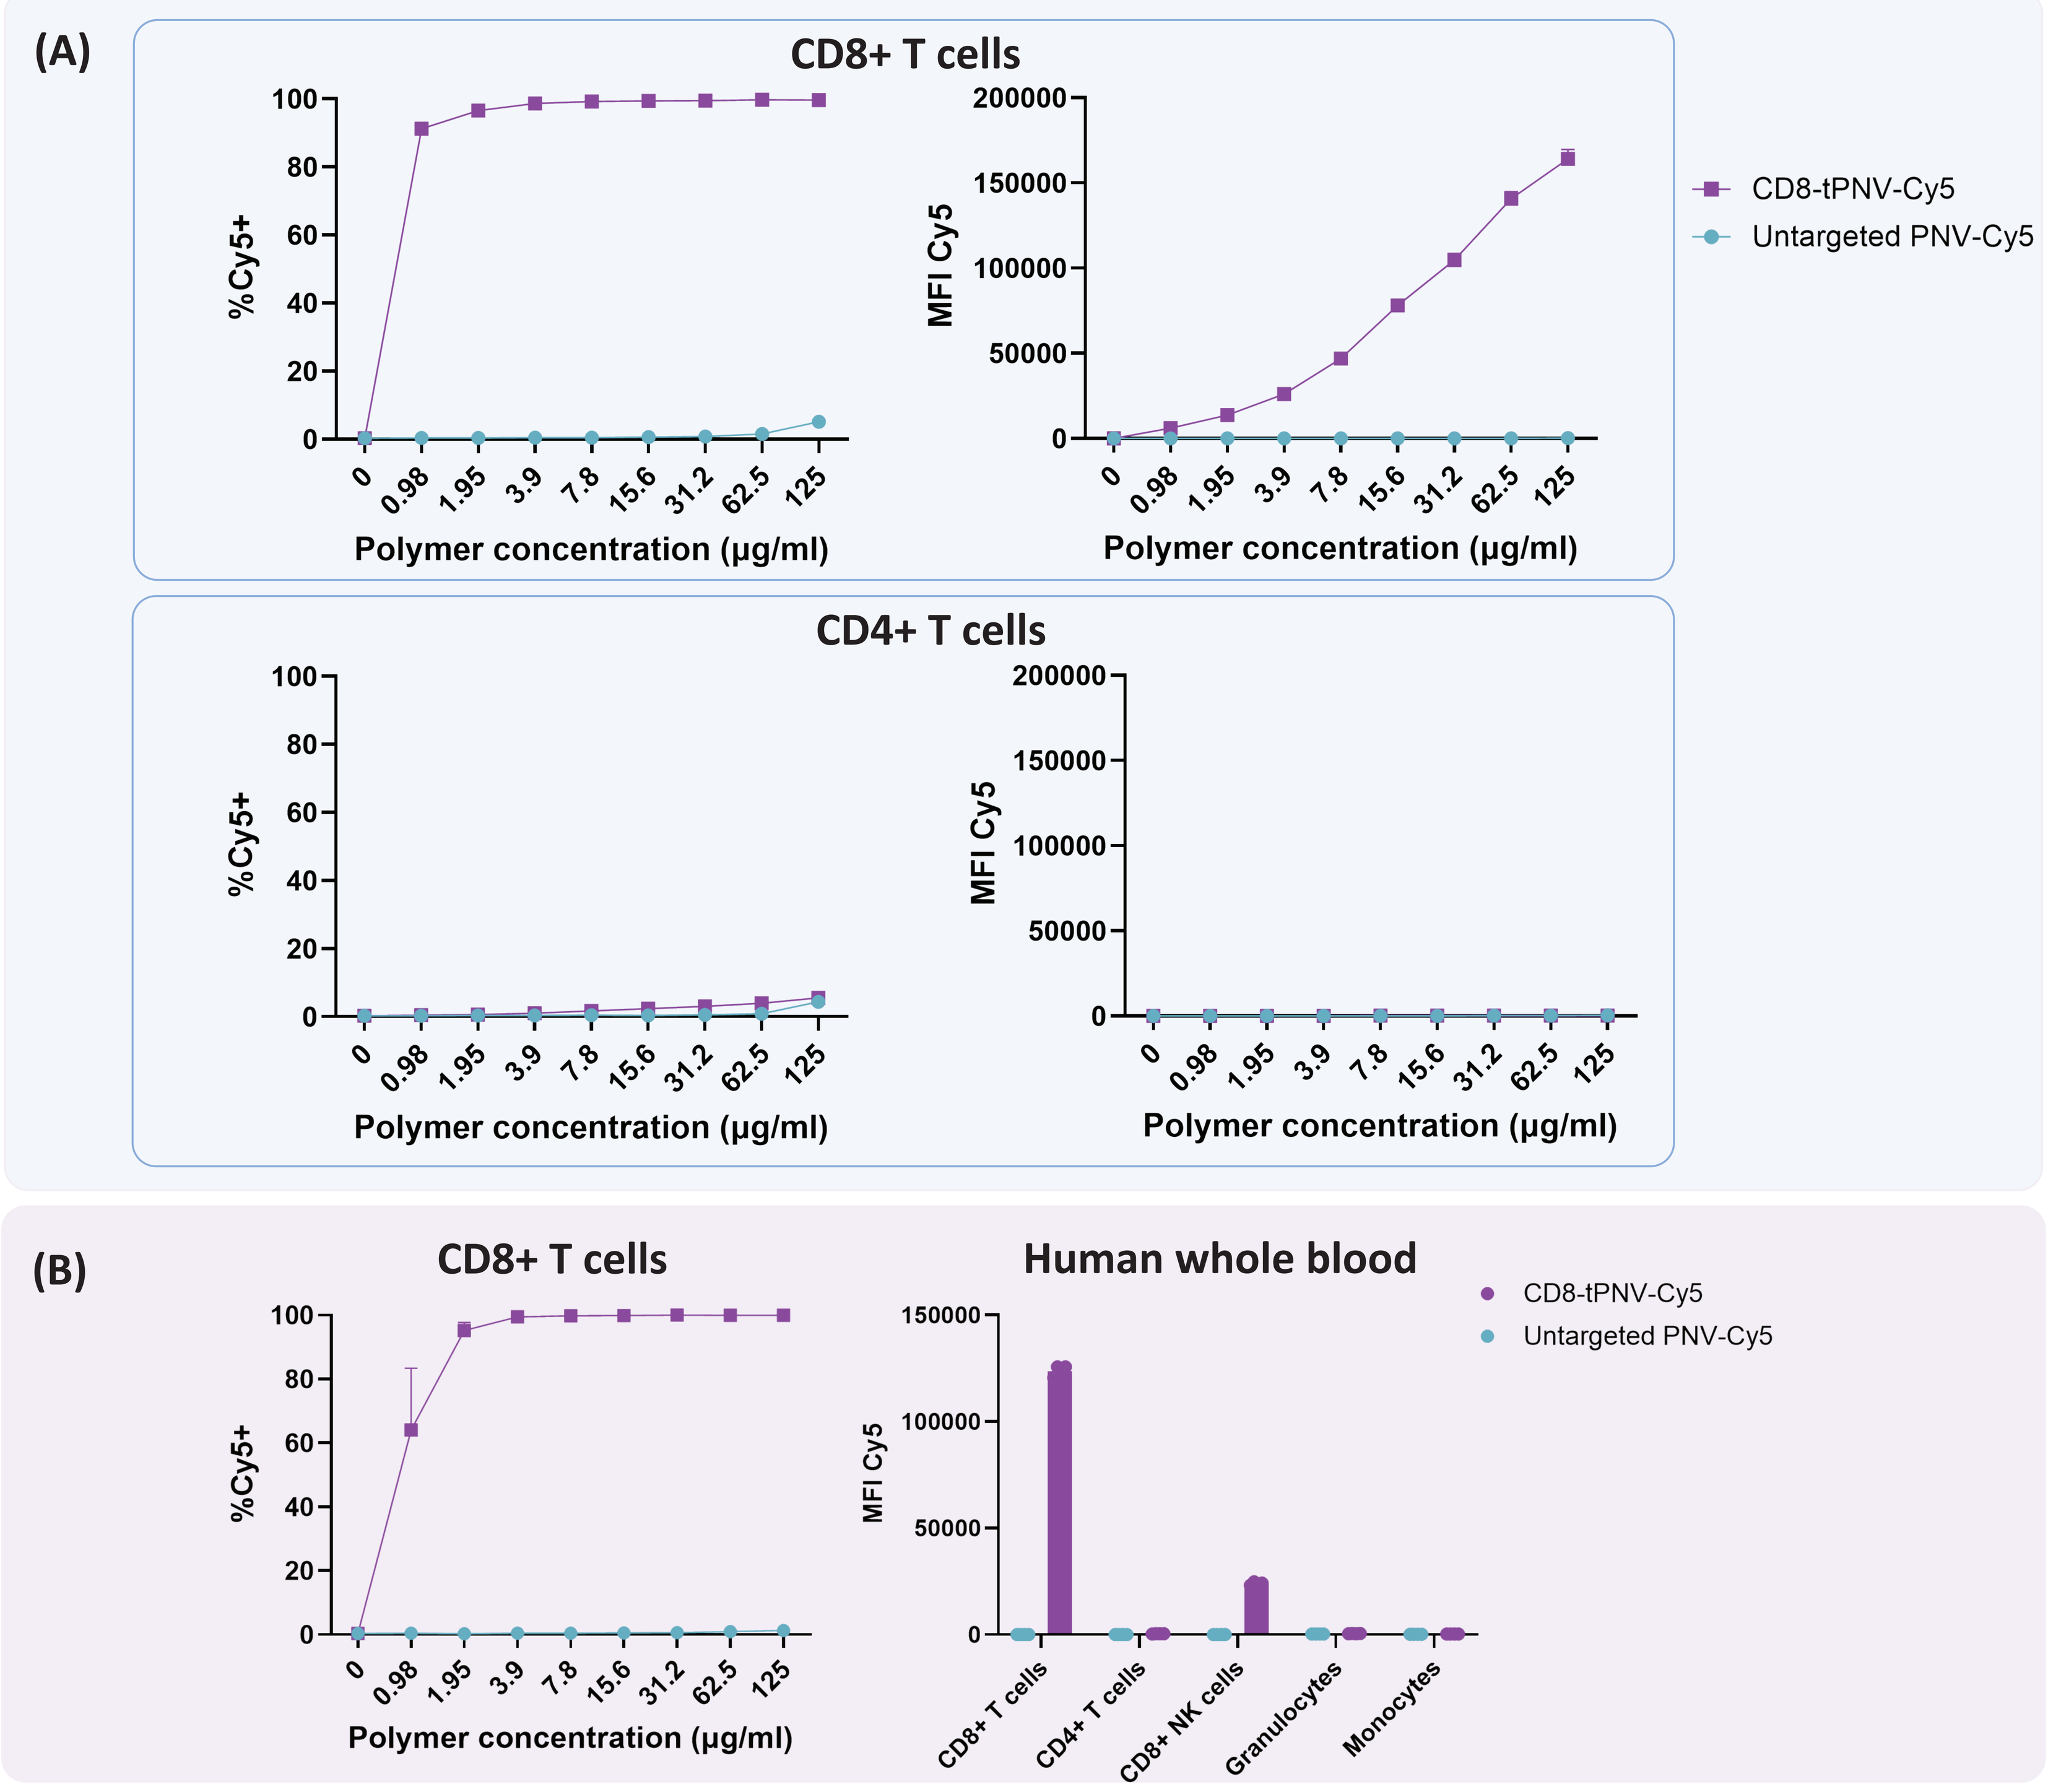
(A)
CD8+ T cells
CD4+ T cells
%Cy5+
MFI Cy5
Polymer concentration (µg/ml)
CD8-tPNV-Cy5
Untargeted PNV-Cy5
(B)
CD8+ T cells
Human whole blood
CD8+ T cells
CD4+ T cells
CD8+ NK cells
Granulocytes
Monocytes

(A) CD8-tPNVs were generated by conjugating VHHs against CD8 to PNVs. Peripheral blood mononuclear cells (PBMCs) from 2 different donors (100,000 cells per well) were treated with increasing concentrations of CD8-tPNVs, incubated for 30 minutes at 37°C. Targeting of CD8+ T cells by CD8-tPNV, but not CD4+ T cells, were observed, while untargeted PNVs showed no CD8 targeting. **(B)** Same CD8 targeting specificity of CD8-tPNVs was observed in human whole blood.

Figure 4. In vivo validation of immune cell targeting by CD8-tPNVs

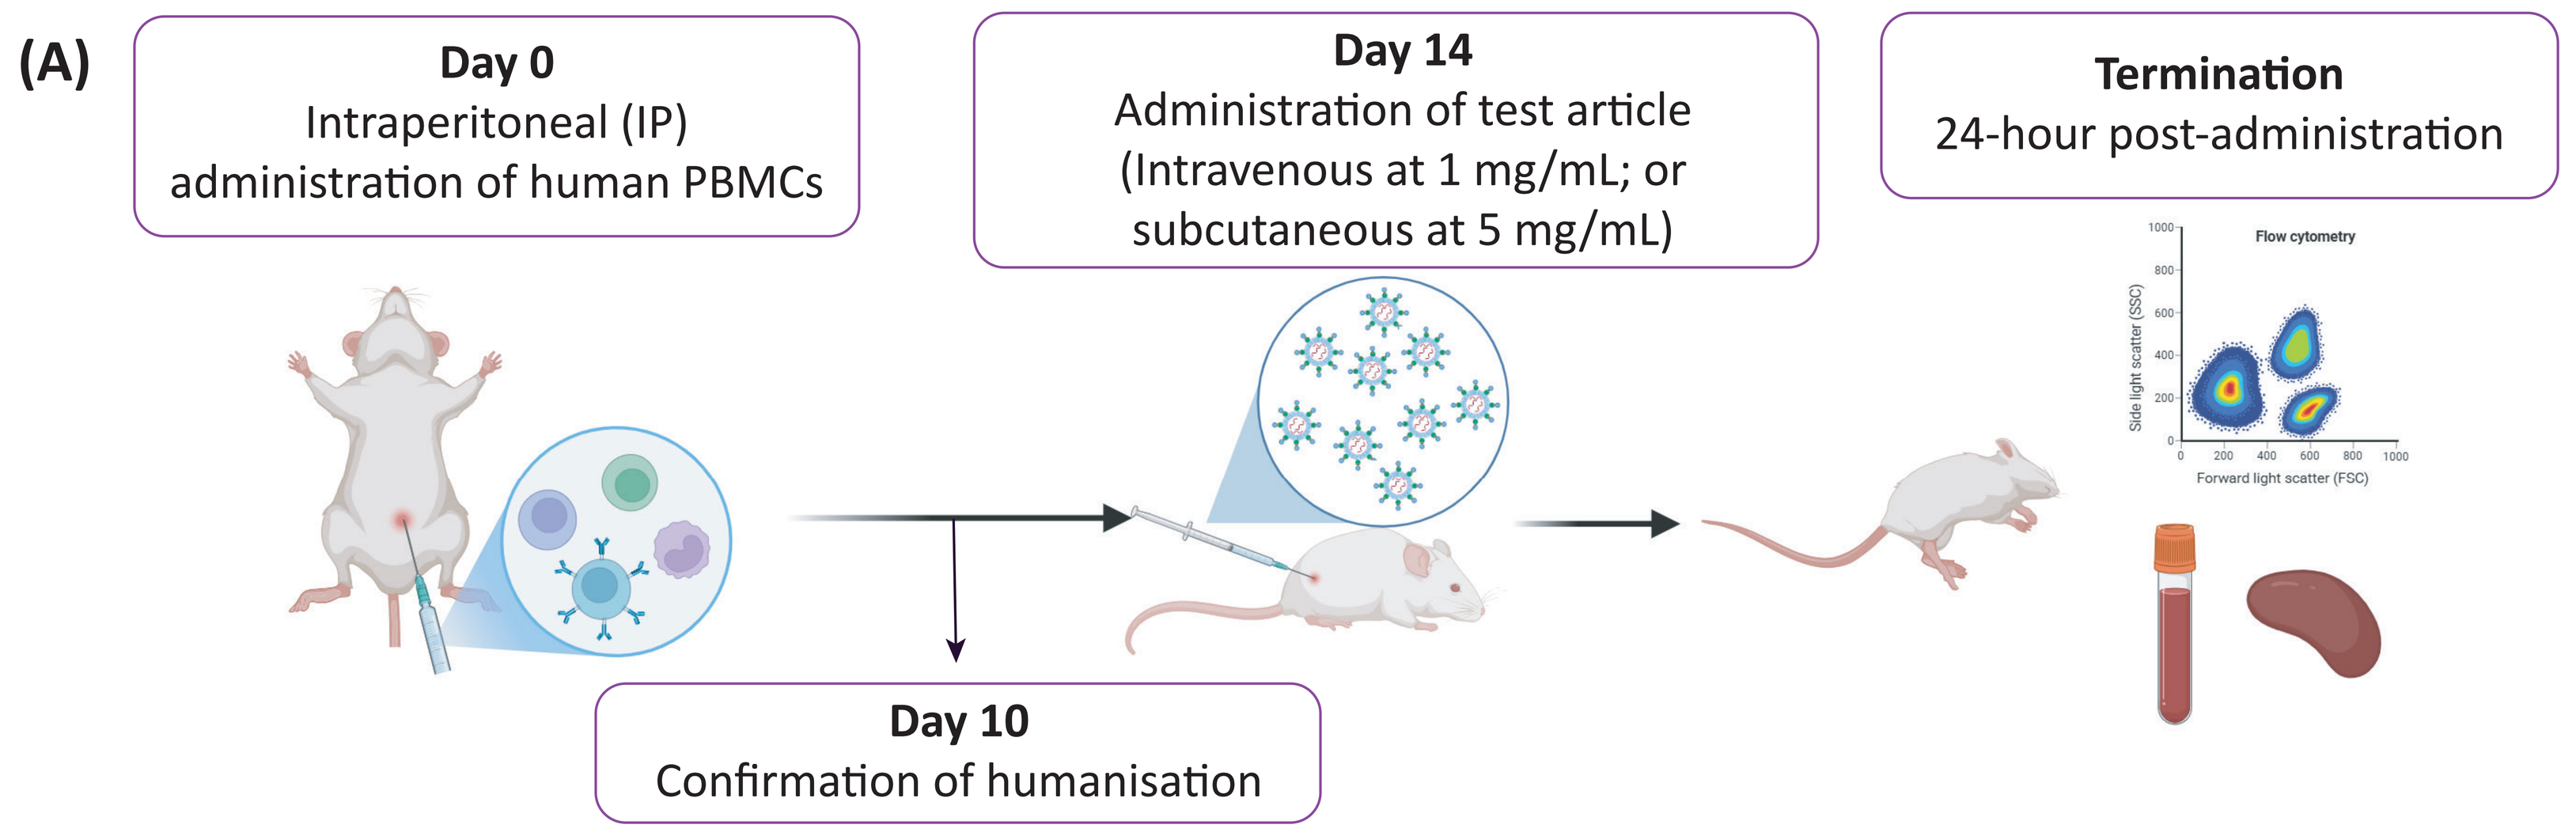
(A)
Day 0
Intraperitoneal (IP) administration of human PBMCs
Day 14
Administration of test article (Intravenous at 1 mg/mL; or subcutaneous at 5 mg/mL)
Termination
24-hour post-administration
Day 10
Confirmation of humanisation
Flow cytometry

(A) Study design for assessing the ability of CD8-tPNVs to target CD8-positive immune cells. Adult NSG mice were inoculated with human PBMCs and humanisation was confirmed at Day 10 by flow cytometry. Test articles (untargeted PNV-Cy5 or CD8-tPNV-Cy5) were administered either intravenously or subcutaneously. After termination, whole blood and spleen samples were collected and analysed using flow cytometry.

Figure 5. In vivo precise immune cell targeting by CD8-tPNVs

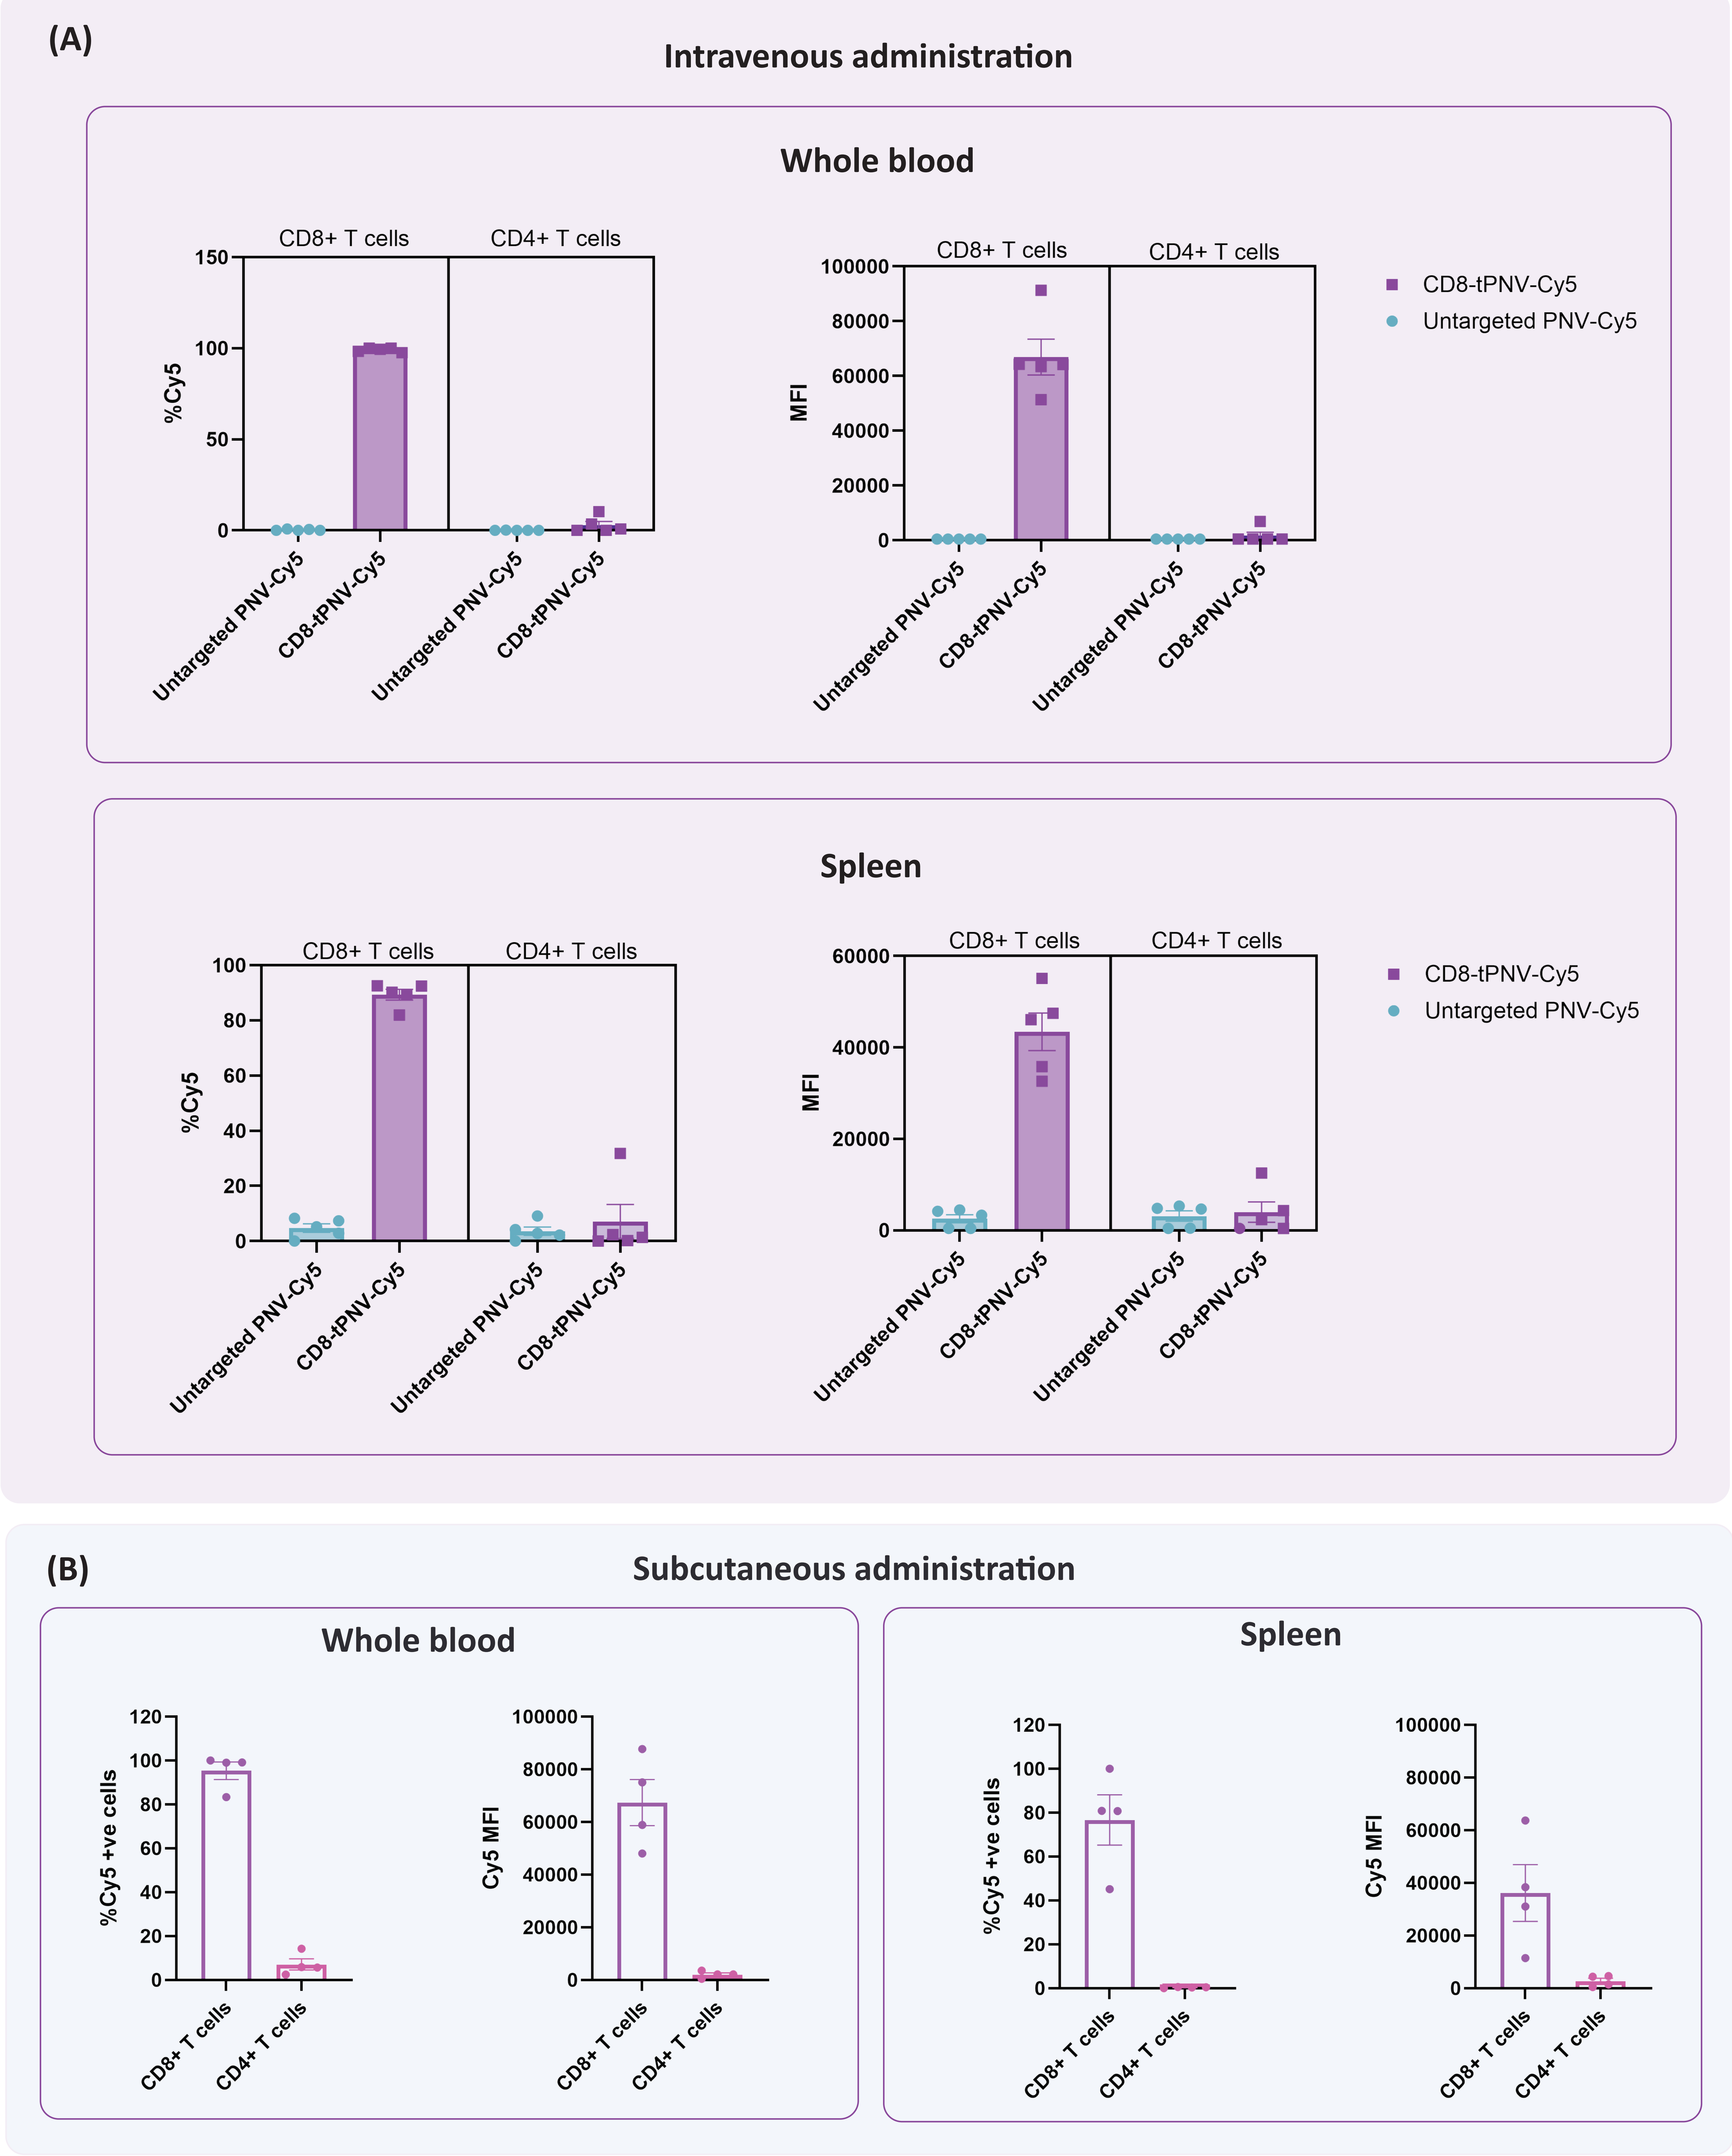
(A)
Intravenous administration
Whole blood
Spleen
CD8+ T cells
CD4+ T cells
%Cy5
MFI
Untargeted PNV-Cy5
CD8-tPNV-Cy5
(B)
Subcutaneous administration
Whole blood
Spleen
%Cy5 +ve cells
Cy5 MFI

(A) Compared to untargeted PNVs, CD8-tPNVs can specifically target CD8+ cells in the whole blood of NSG humanised PBMC mice through intravenous administration. In the spleen, more than 80% of CD8+ T cells were detected with CD8-tPNV-Cy5, indicating deep tissue penetration, which is critical for 'immune reset' in autoimmune disease settings. **(B)** CD8 cell-specific targeting of CD8-tPNVs was also observed in whole blood and spleen of mice dosed subcutaneously with CD8-tPNVs. Subcutaneous administration may provide a more cost-effective and easy to self-administer route in treatments that require repeat dosing.

Conclusions

- tPNVs, comprised of three key components, offer a versatile, plug-and-play polymer-based platform for the delivery of oligonucleotides.
- tPNVs are proprietary and redosable, with no immunogenicity observed.
- tPNVs are simple to manufacture with controllable and reproducible product attributes.
- tPNVs enable specific delivery to any cells and tissues.
- CD8-tPNVs precisely target CD8-expressing immune cells in vitro and in vivo after intravenous or subcutaneous administration.
- tPNVs can be developed broadly across areas such as I&I, Oncology, CNS, respiratory and other diseases.